NativeSkin®, an immunocompetent human skin model to study antigen uptake and processing by Langerhans cells

Pagès E.[1], Mondoulet L.[2], Bonnefont E.[1], Braun E.[1], Dhelft V.[2], Dupont C.[3], Sampson H.[4], Descargues P.[5]

[1]Genoskin, Toulouse, France (contact@genoskin.com) ; [2]DBV Technologies, Research & Innovation Dpt, Montrouge, France ; [3]Hôpital Necker, Université Paris-Descartes, Paris, France ;
[4]DBV Technologies, Research & Innovation Dpt, New York, USA [5]Genoskin Inc. Boston (MA), USA

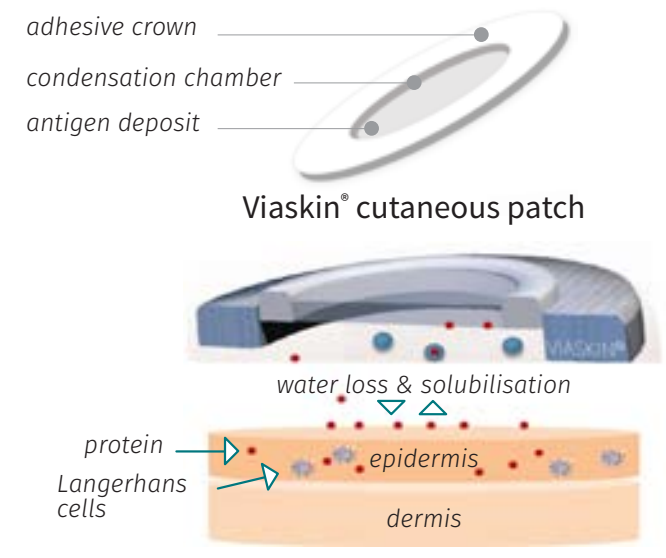
adhesive crown
condensation chamber
antigen deposit
Viaskin® cutaneous patch
water loss & solubilisation
protein
Langerhans cells
epidermis
dermis
VIASKIN® SCHEMATIC

Introduction

Allergen applied to the skin during epicutaneous immunotherapy (EPIT) with Viaskin® patches seemed to induce tolerance in sensitized mice (Dioszeghy, Clin Exp Allergy 2014). Safety and adequate dose for treatment with Viaskin® Peanut in peanut allergic patients was studied in earlier Phase 1 and Phase 2 clinical trials and completed Phase 3 safety and efficacy trial. In preliminary studies in sensitized mice, DBV Technologies showed that allergen delivered by Viaskin® is mainly taken-up by Langerhans cells (LCs) and transported to regional lymph nodes.

THE AIM OF THIS STUDY WAS TO DETERMINE WHETHER SIMILAR ALLERGEN UPTAKE OCCURRED IN AN *EX VIVO* HUMAN SKIN MODEL.

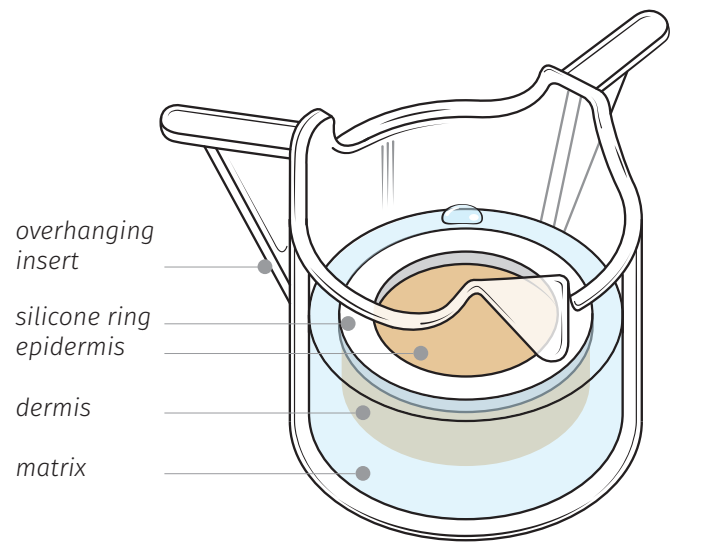
overhanging insert
silicone ring
epidermis
dermis
matrix
NATIVESKIN® SCHEMATIC

Experimental workflow

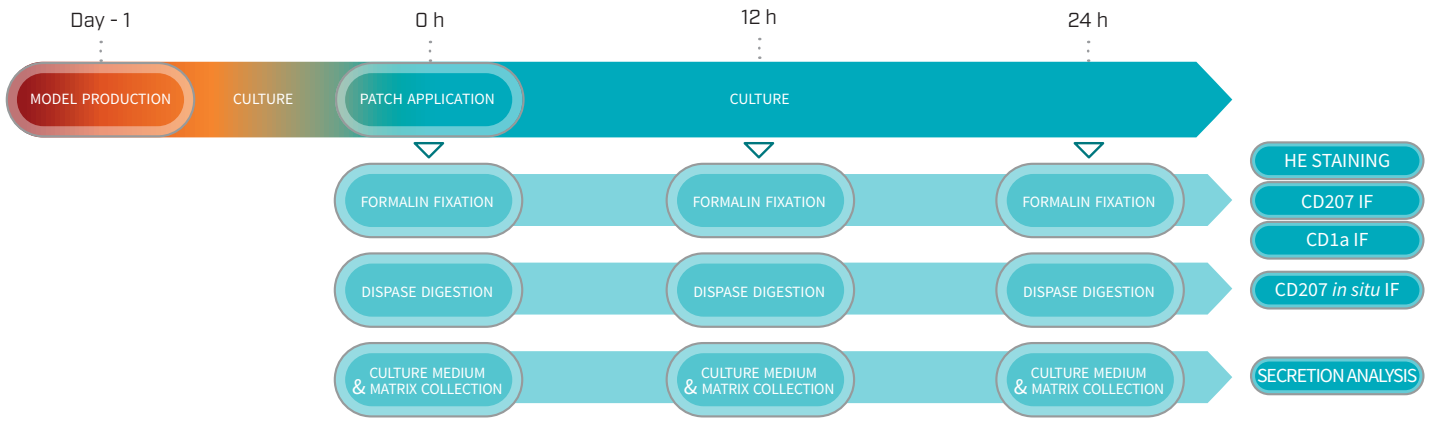
Day - 1
0 h
12 h
24 h
MODEL PRODUCTION
CULTURE
PATCH APPLICATION
CULTURE
FORMALIN FIXATION
DISPASE DIGESTION
CULTURE MEDIUM & MATRIX COLLECTION
HE STAINING
CD207 IF
CD1a IF
CD207 in situ IF
SECRETION ANALYSIS

TREATMENT WITH VIASKIN PLACEBO VS VIASKIN PEANUT (A-647) DURING 24H
The experiment was performed on 2 donors and in triplicate by condition. Viaskin® patches were applied on the skin after the production of NativeSkin® models.

Skin integrity following Viaskin patch application during 24 hours

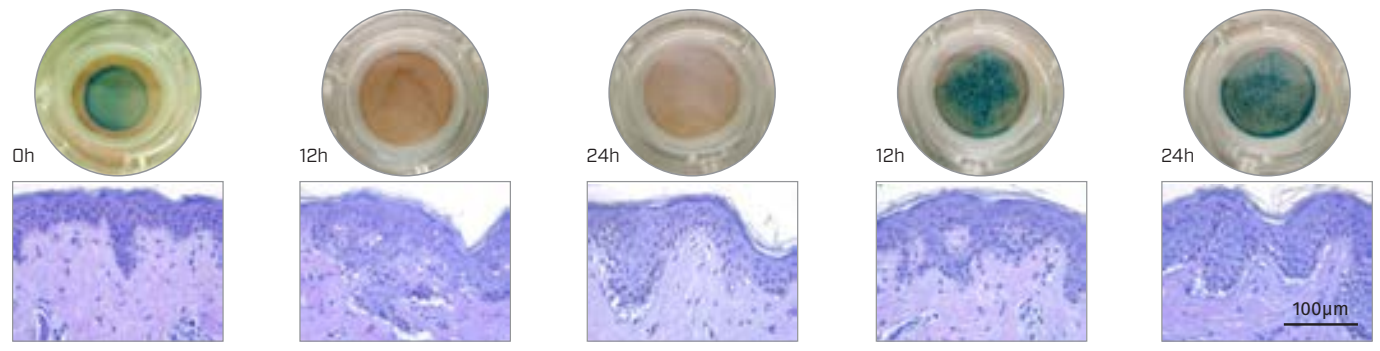
Viaskin Placebo
Viaskin Peanut
0h
12h
24h
12h
24h
100µm

MACROSCOPICAL AND HISTOLOGICAL EVALUATION OF TISSUE INTEGRITY DURING TREATMENT
For each condition, representative pictures of models and the epidermis/dermis sections were imaged. Results for donor 2 are shown and are similar to those obtained for donor 1. On macroscopical pictures, it is possible to observe allergen solubilization due to perspiration.

NO MAJOR ALTERATION OF THE SKIN AFTER VIASKIN PATCHES APPLICATION DURING 24H

Antigen penetration in the epidermis on skin cross sections

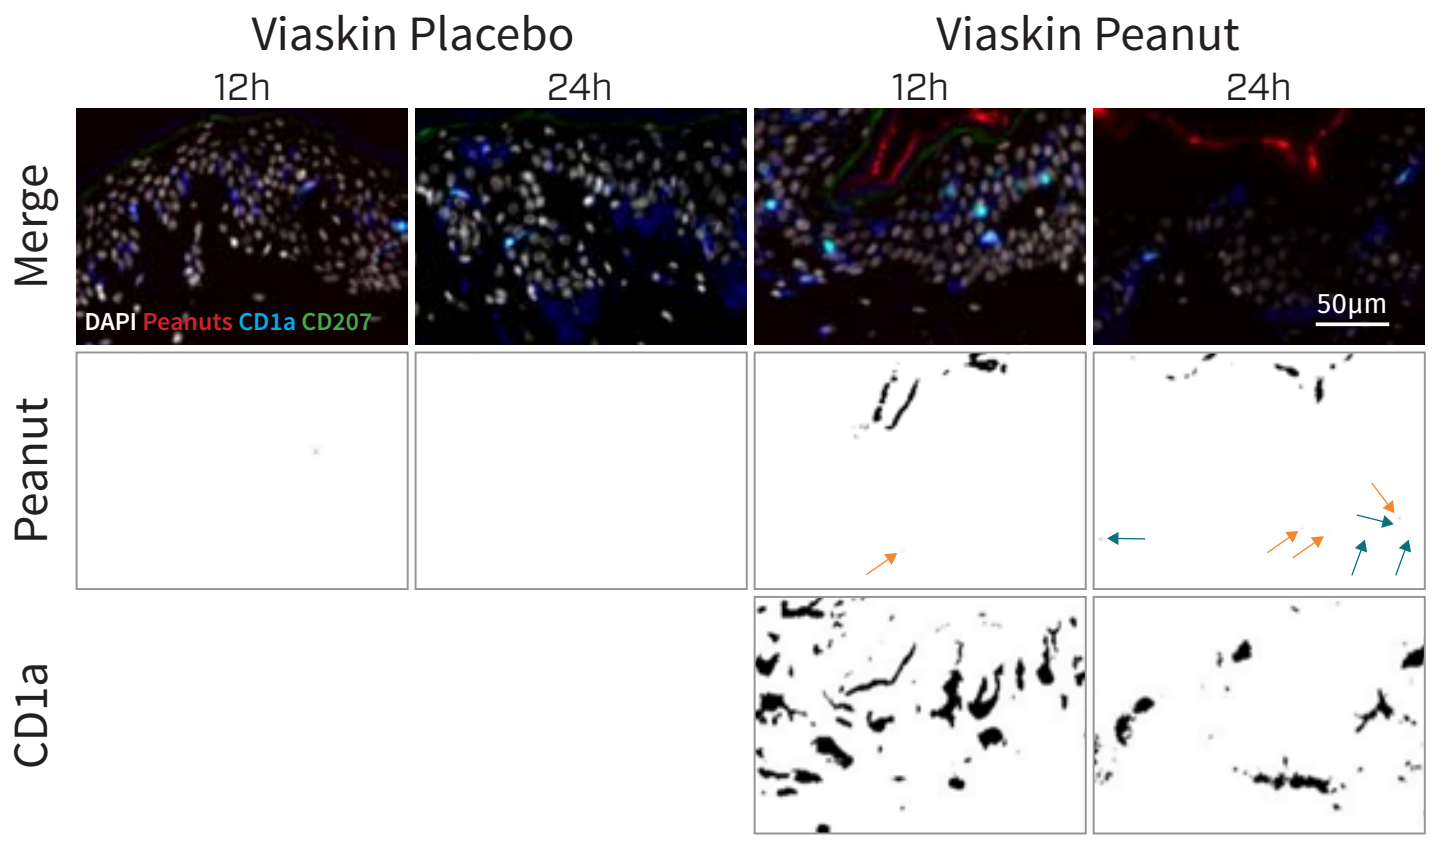
Viaskin Placebo
Viaskin Peanut
12h
24h
12h
24h
Merge
Peanut
CD1a
DAPI Peanuts CD1a CD207
50µm

STUDY OF ALLERGEN PENETRATION AND COLOCALIZATION WITH LANGERHANS CELLS ON TISSUE SECTIONS
Representative pictures for donor 2, on merged pictures, cell nuclei stained with DAPI (Grey), LC with anti-CD1a (Blue) and anti-CD207 (Green) and peanut (Red). On Peanut pictures, allergen particles appear in black, blue arrows point Peanut in the epidermis, and orange arrows target allergen colocalizing with CD1a positive cells (visible in black on the last line).

ALLERGEN PENETRATION IN THE EPIDERMIS & COLOCALIZATION WITH LANGERHANS CELLS

QUANTIFICATION OF ALLERGEN PENETRATION AND COLOCALIZATION WITH LANGERHANS CELLS
Peanut in the epidermis (Blue) and colocalization with LC (orange) were detected using Fiji for donors 1 and 2 on skin cross sections. Results are presented for Viaskin Peanut. The analysis was performed for Viaskin Placebo, no Peanut signal was detected.

VARIABLE KINETICS OF PENETRATION BETWEEN DONORS

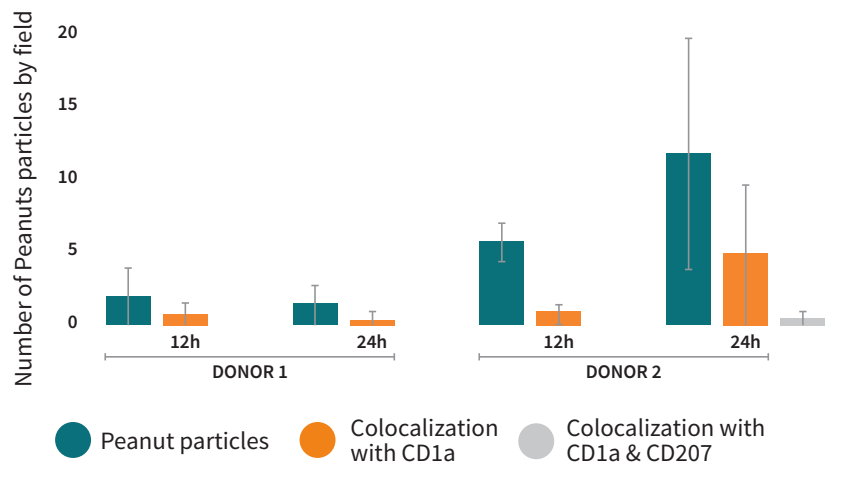
Number of Peanuts particles by field
20
15
10
5
0
12h
24h
12h
24h
DONOR 1
DONOR 2
Peanut particles
Colocalization with CD1a
Colocalization with CD1a & CD207

Conclusion

NativeSkin® model is a relevant model to investigate the local immune response related to a skin treatment. It allows to confirm the role of LCs in the antigen uptake and processing after Viaskin® application on human skin. Particularly, we showed that Viaskin® Peanut applied on *ex vivo* human skin leads to the antigen penetration in the epidermis and its uptake by Langerhans cells within 24 hours (with variable kinetics depending on the donor).

Colocalization of allergen-Langherans cells by *in situ* immunostaining

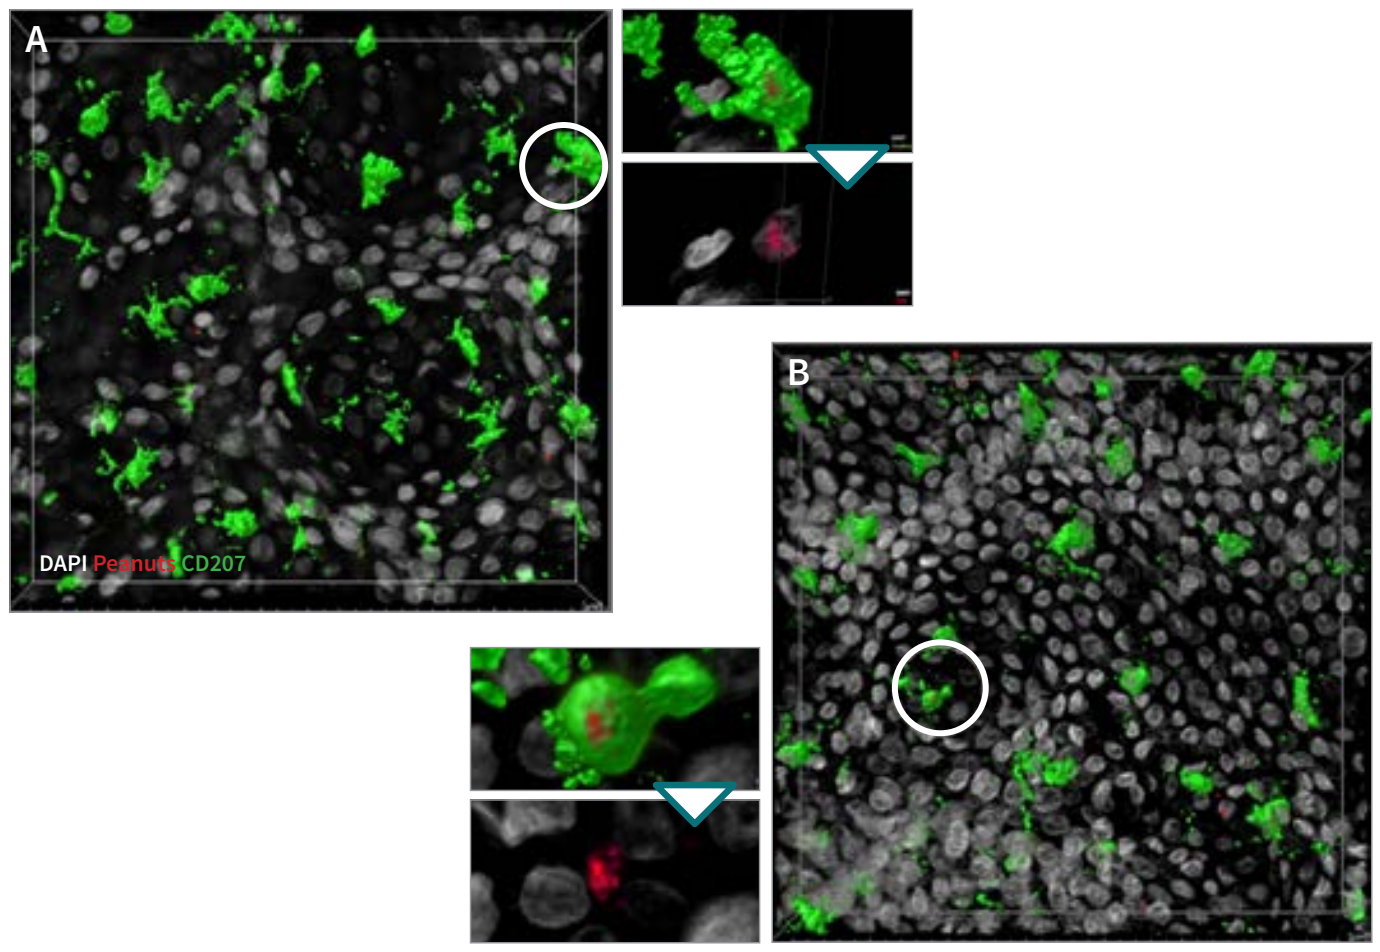
A
B
DAPI Peanuts CD207

OBSERVATION OF *IN SITU* IMMUNOSTAINING ON DETACHED EPIDERMAL SHEET AFTER 24H OF TREATMENT WITH VIASKIN PEANUT USING CONOFCAL MICROSCOPY
Representative pictures for donor 1 (A) and donor 2 (B) of penetration of peanut in Langerhans cells positive for *in situ* CD207 immunostaining of the epidermis of NativeSkin® models treated with Viaskin® Peanut during 24 hours. Global view of 3D reconstruction. White circles allow to localize peanut and CD207 positive cells. In the center, detailed view of positive Langerhans cells after 24 hours of treatment. CD207 signal was removed to visualize internalization of antigen.

COLOCALIZATION OF ALLERGEN WITH LANGERHANS CELLS AFTER VIASKIN TREATMENT

Cytokines release after Viaskin application

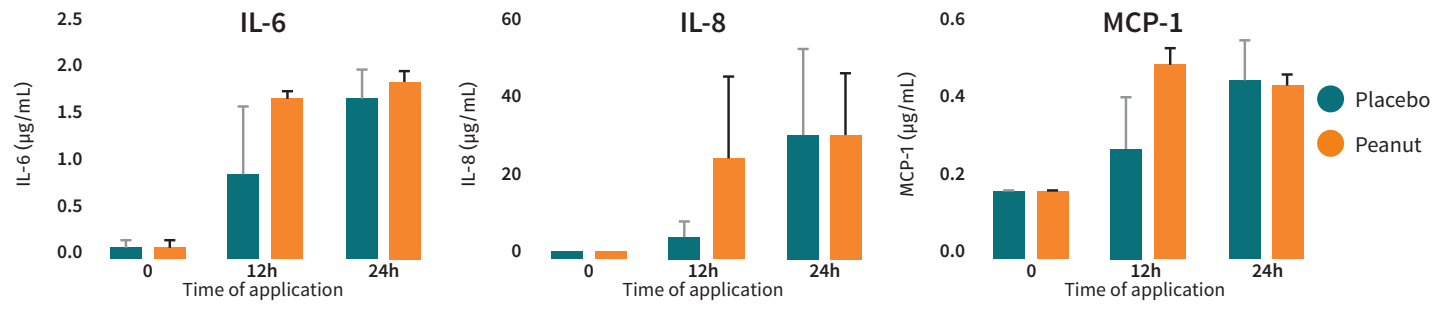
IL-6
IL-6 (µg/mL)
2.5
2.0
1.5
1.0
0.5
0.0
0
12h
24h
Time of application
IL-8
IL-8 (µg/mL)
60
40
20
0
MCP-1
MCP-1 (µg/mL)
0.6
0.4
0.2
0.0
Placebo
Peanut

MEASUREMENTS OF CYTOKINES RELEASED IN THE CULTURE MEDIUM DURING 24H OF TREATMENT WITH VIASKIN
Cytokines detected in culture medium (after 24 hours of treatment with Viaskin®). A human 27-Plex® kit (Bio-plex®, Biorad) was used for the quantitative detection of cytokine in culture medium. Only 3 cytokines were detected above the limit of quantification: IL-6, IL-8, MCP1. Results are presented for donor 1 and are similar for donor 2. The bars represent the mean ± SD of triplicates.

AFTER VIASKIN APPLICATION, SIMILAR DETECTION OF CYTOKINES BETWEEN PEANUT AND PLACEBO COULD EXPLAIN THE ABSENCE OF INFLAMMATION OCCURING DURING TREATMENT